Mimicking molecular conversation by modulation of the plasminogen serine protease system in order to improve skin barrier function.

Eileen Jackson [1], Rainer Voegeli[1], Peter Wikstroem[1], Anthony V. Rawlings[2]

[1]DSM Nutritional Products Ltd., Personal Care, Kaiseraugst; [2]AVR Consulting Ltd., Northwich, UK

* correspondance to: eileen.jackson@dsm.com

Summary

BSFAB (INCI name: Benzylsulfonyl-D-Seryl-Homophenylalanine Amidino-Benzylamide Acetate) (Figure 1), a dual inhibitor of plasmin and urokinase, two important components of the plasminogen system, has been shown to improve skin barrier function. In this study the mode of binding of BSFAB in plasmin and urokinase was predicted based on existing crystal structures and molecular modeling tools. The resulting *in silico* model helps to rationalize the key protein ligand interactions that are responsible for the dual inhibitory action of this dipeptide derivative.

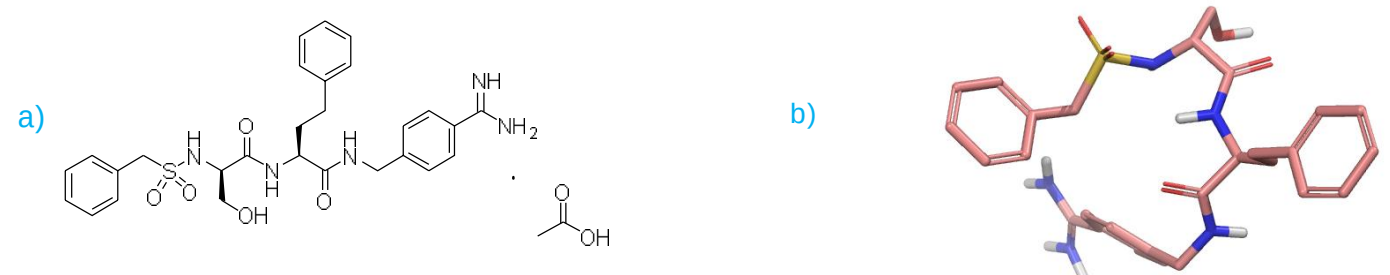
a)
b)

Figure 1. *Chemical structure of BSFAB, a) 2D structure; b) 3D structure predicted for binding site*

Background

Plasmin and urokinase are trypsin-like serine proteases that are upregulated in barrier compromised skin and are thought to be the major protease activity involved in the delay of barrier recovery.[1,2] A reversible, competitive inhibitor that selectively targets these two enzymes has the potential to improve barrier function and is thus highly desirable.

Plasmin, and urokinase both possess a two-domain structure with a ß-barrel in each domain and a catalytic triad consisting of histidine, aspartic acid and serine.[3]

Also characteristic is a deep S1 primary or specificity binding pocket with an acidic Asp at the bottom, which forms a salt bridge with the basic Arg or Lys as P1 residues in endogenous substrates. Existing X-ray crystal structures for both plasmin and urokinase catalytic domains (Figure 2) allow the use of computational approaches to 'dock' small molecules into the protein binding site thus predicting the presumable optimal ligand conformation and orientation within its biological target.

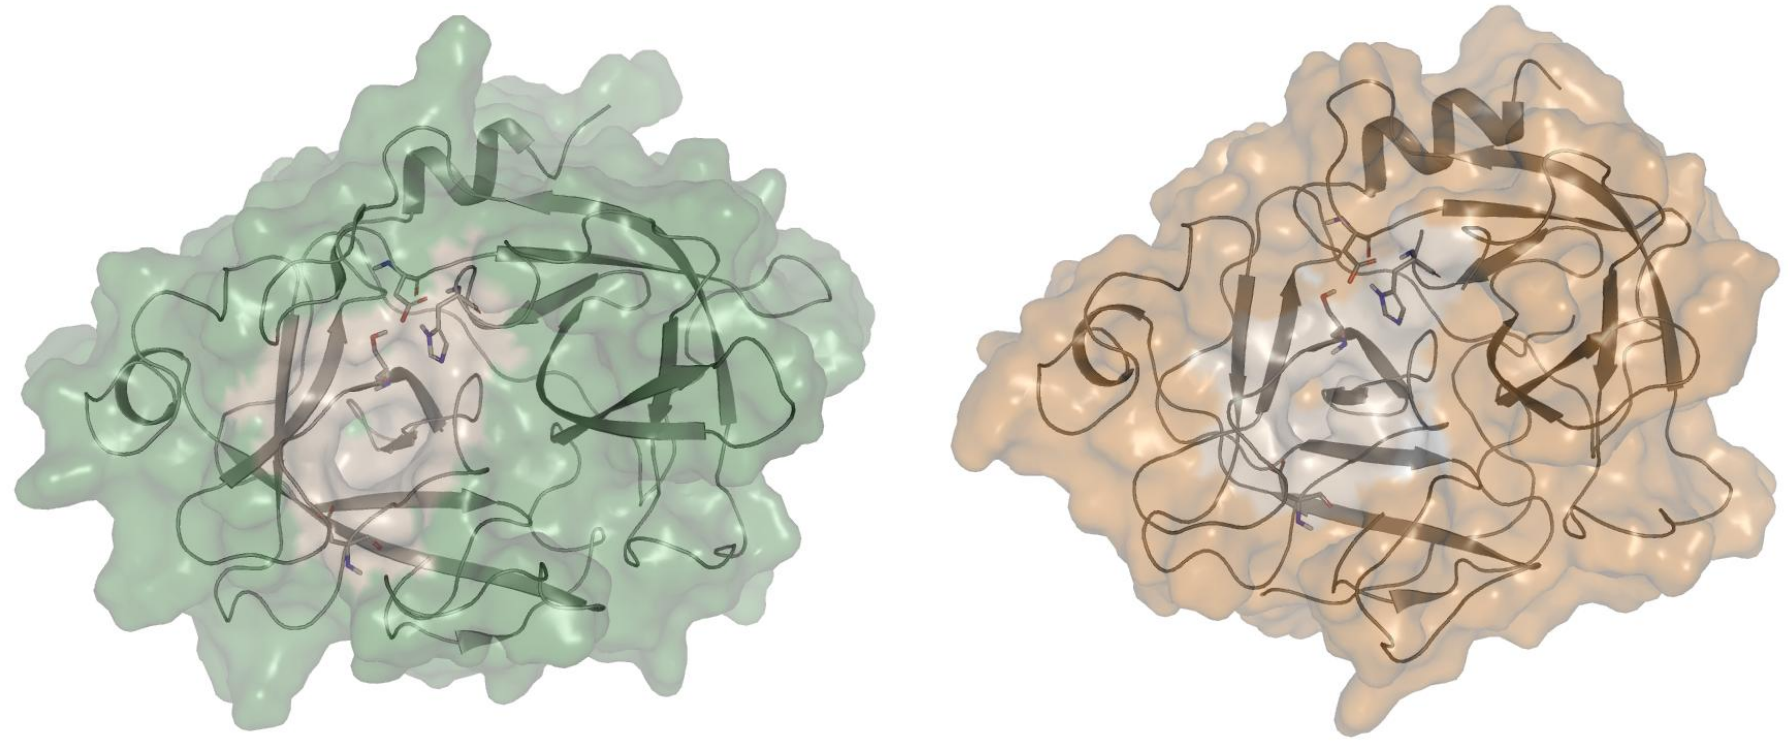

Figure 2*. X-ray crystal structure of catalytic domain of urokinase (left) and plasmin (right), highlighting binding pocket*

Methods

The catalytic domain of plasmin and urokinase were retrieved from the protein data bank of Research Collaboration for Structural Bioinformatics (RSCB PDB) (codes: 1BUI and 1VJ9, respectively) and prepared for modelling studies using a protein preparation wizard (Maestro version 10.4, Schrödinger, LLC, New York, NY, 2015). BSFAB was docked to the catalytic domains (Glide, version 6.9, Schrödinger, LLC, New York, NY, 2015) and the Standard Precision (SP) setting was used. For docking, multiple conformers of BSFA were generated (Confgen, version 3.1, Schrödinger, LLC, New York, NY, 2015). Among the different binding poses generated the best pose, based on visual inspection, was refined using Prime, version 4.2, Schrödinger, LLC, New York, NY, 2015, Figures 1b,2,3 were prepared using the PyMOL Molecular Graphics System, Version 1.8.0.4 Schrödinger, LLC.

Results

The mode of binding of a highly active and specific dual competitive inhibitor was rationalized by docking into plasmin and urokinase (Figure 3). In both cases BSFAB adopts a turn-like conformation with its peptide backbone forming a short antiparallel ß-sheet with residues Ser214 and Gly216. The guanidine amidine moiety of BSFAB is tightly anchored into the S1 pocket by a salt bridge with the sidechain of Asp 189 as well as H-bond interactions with the carbonyl group of Gly219 and the hydroxyl sidechain of Ser190. Another common interaction in both complexes is the H-bond between the NH of Gly219 and one oxygen atom of BSFAB sulfonamide group.

The urokinase catalytic domain has an extra specific insertion loop that is not present in plasmin. The D-Ser residue of BSFAB is located next to this loop and its sidechain hydroxyl forms extra H-bond interactions with the carbonyl oxygen of Leu97B and imidazole side chain of His99. Another small difference is that plasmin forms a H-bond interaction with the sidechain of Gln192 that is rotated away in the urokinase structure and thus not observed. This glutamine residue is known to be highly flexible adopting different orientations in different complexes.

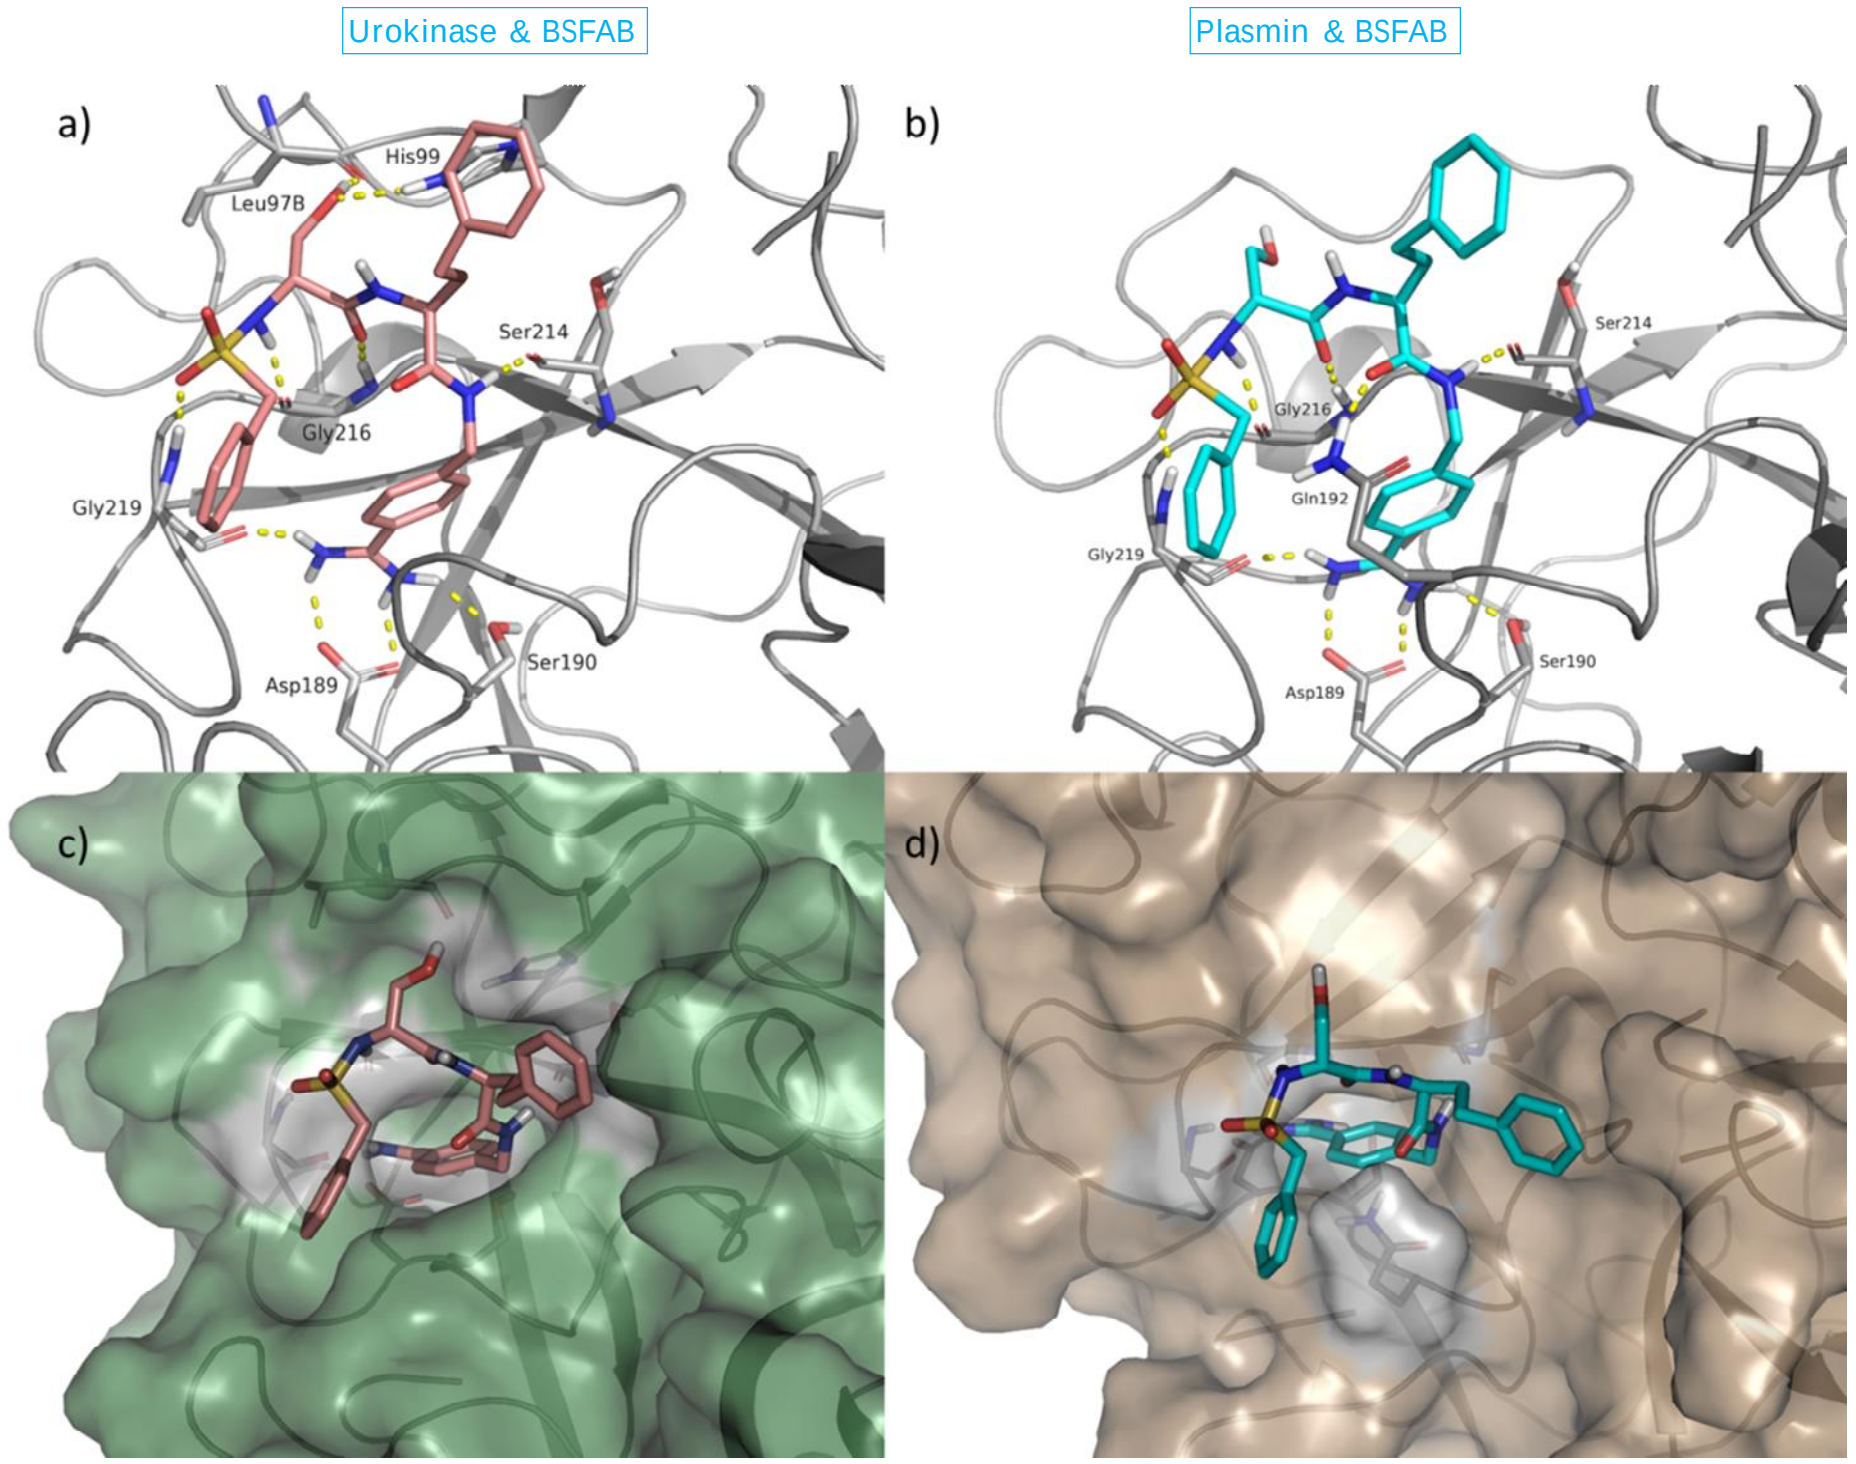
Urokinase & BSFAB
Plasmin & BSFAB
a)
His99
Leu97B
Ser214
Gly216
Gly219
Asp189
Ser190
b)
Ser214
Gly216
Gln192
Gly219
Asp189
Ser190
c)
d)

Figure 3*. Modeled binding modes of BSFAB. Cartoon view of BSFAB bound into urokinase (a) and plasmin (b) highlighting key hydrogen bond interactions that result both in a short antiparallel β-sheet and tight binding into the S1 pocket. Surface view of BSFAB bound into urokinase (c) and plasmin (d) highlighting the fit of BSFAB into the S1 pocket and the main contact residues.*

29th IFSCC Congress, 2016, Orlando

References

1. Voegeli, R.; Rawlings, A. V.; Doppler, S.; Heiland, J.; Schreier, T. *Int. J. Cosmet. Sci,* 29 (3), 191–200, *2007*.
2. Voegeli, R.; Rawlings, A. V.; Doppler, S.; Schreier, T. *Int. J. Cosmet. Sci*. 30 (6), 435–442, *2008*.
3. Liang, G.; Bowen, *J. Curr. Top. Med. Chem*. 16 (13), 1506–1529, *2016*.

Acknowledgements

This study was fully funded by DSM Nutritional Products Ltd, Basel, CH